156

Tucatinib combined with trastuzumab and pertuzumab as first-line maintenance therapy for HER2+ metastatic breast cancer: An in-depth safety analysis of HER2CLIMB-05

Veronique Dieras,[1] Giuseppe Curigliano,[2,3] Miguel Martin,[4] Florence Lerebours,[5] Junji Tsurutani,[6] Marie-France Savard,[7] Katarzyna J. Jerzak,[8] Xichun Hu,[9] Luciana Carla Martins de Aquino Pimentel,[10] Ciara C. O'Sullivan,[11] Eriko Tokunaga,[12] Alicia Okines,[13,14] Chiun-Sheng Huang,[15] Joohyuk Sohn,[16] Eduardo Cronemberger Silva,[17] Volkmar Mueller,[18] Manjot Kaur,[19] Helene Viala,[20] Shan Yang,[21] Erika Hamilton[22]

[1]Centre Eugène Marquis, Rennes, France; [2]European Institute of Oncology, IRCCS, Milano, Italy; [3]University of Milano, Milano, Italy; [4]Hospital General Universitario Gregorio Marañon, Universidad Complutense, CIBERONC, GEICAM, Madrid, Spain; [5]Institut Curie, Saint-Cloud, Paris, France; [6]The Innovative Center of Translational Research and Clinical Science for Cancer Therapy, Showa Medical University Hospital, Tokyo, Japan; [7]The Ottawa Hospital Cancer Centre, Ottawa, Ontario, Canada; [8]Sunnybrook Odette Cancer Centre, University of Toronto, Toronto, Ontario, Canada; [9]Fudan University Shanghai Cancer Center, Shanghai, China; [10]Liga Norte-Rio Grandense Contra o Cancer, Brazil; [11]Mayo Clinic, Rochester, MN, USA; [12]National Hospital Organization Kyushu Cancer Center, Fukuoka, Japan; [13]The Royal Marsden NHS Foundation Trust, London, UK; [14]Institute of Cancer Research, London, UK; [15]Breast Care Center, National Taiwan University Hospital, Taipei, Taiwan; [16]Yonsei Cancer Center, Yonsei University Health System, Seoul, Republic of Korea; [17]Centro Regional Integrado de Oncologia (CRIO), Fortaleza, Brazil; [18]University Breast Center, Universitaetsklinikum Hamburg-Eppendorf, Hamburg, Germany; [19]Pfizer Inc, Kirkland, Quebec, Canada; [20]Pfizer International Operations, Paris, France; [21]Pfizer Inc, Bothell, WA, USA; [22]Sarah Cannon Research Institute, Nashville, TN, USA

Objective

To report the findings from an in-depth safety analysis of the phase 3 HER2CLIMB-05 study evaluating the efficacy and safety of tucatinib (TUC) added to first-line (1L) maintenance therapy with trastuzumab (H) and pertuzumab (P) in patients with human epidermal growth factor receptor 2-positive (HER2+) metastatic breast cancer (MBC).

Conclusions

In HER2CLIMB-05, hepatic adverse events (AEs) and diarrhea were more frequently seen in the TUC arm compared with the placebo (PBO) arm. The majority of hepatic AEs were elevated liver enzymes, which were mostly asymptomatic, transient, and manageable with dose modifications and/or discontinuations, with no severe outcomes observed. Most diarrhea AEs were of low severity and managed with antidiarrheals and/or dose modifications.

TUC addition to 1L HP maintenance therapy represents an effective option for HER2+ MBC with a clinically manageable safety profile.

Lead author: Veronique Dieras

Email for more information: v.dieras@rennes.unicancer.fr

Poster **aPLS**

An electronic version of this poster and abstract in plain language may be obtained by scanning this Quick Response (QR) code with your smartphone app. Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

References: 1. Genentech, Inc. Highlights of prescribing information. Perjeta. April 2025. **2.** Gion M, Trapani D, Cortés A, et al. *Am Soc Clin Oncol Educ Book*. 2022;42:1-11. **3.** Baselga J, Cortés J, Kim SB, et al. *N Engl J Med*. 2012;366:109-19. **4.** Swain SM, Miles D, Kim SB, et al. *Lancet Oncol*. 2020;21:519-530. **5.** Dieras V, Giuseppe C, Martin M, et al. *J Clin Oncol*. Dec 2025. doi: 10.1200/JCO-25-02600.

Acknowledgments: Medical writing was provided by Melissa Lingohr-Smith, PhD, of Oxford PharmaGenesis Inc. (Wilmington, DE, USA), and was funded by Pfizer Inc.

Sponsor: This study was sponsored by Seagen, which was acquired by Pfizer in December 2023.

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting
May 29–June 2, 2026 • Chicago, IL, USA

Introduction

- Since approval in 2012,[1] the standard-of-care for 1L treatment of HER2+ MBC has been induction therapy with a taxane (T) in combination with dual anti-HER2 monoclonal antibodies (HP) followed by maintenance therapy with HP until disease progression.[2]
- Treatment with 1L THP induction therapy and HP maintenance therapy significantly prolonged progression-free survival (PFS) in patients with HER2+ MBC,[3] as well as increased survival rate,[4] but most patients eventually experience disease progression within approximately two years of initiating treatment.[4]
- In the primary HER2CLIMB-05 analysis, addition of TUC to 1L maintenance HP in patients with HER2+ MBC extended PFS (per investigator) vs control treatment by nearly 9 months (24.9 vs 16.3 months, respectively; hazard ratio: 0.641, $P < 0.0001$).[5]
 - TUC combined with HP had a tolerable safety profile consistent with the known components of this combination, except for a higher rate of elevated liver transaminases (grade ≥3 events), which were typically manageable and reversible with dose modifications and/or discontinuations.[5]

Methods

TRIAL DESIGN AND PATIENTS

- HER2CLIMB-05 is an ongoing randomized, double-blind, placebo-controlled, international, phase 3 trial (NCT05132582; **Figure 1**).

TREATMENT

- Following induction therapy, patients were randomly assigned 1:1 to TUC or placebo BID, both in combination with maintenance HP.
- Up to 3 dose reductions of TUC or PBO in 50 mg increments (300 mg → 250 mg → 200 mg → 150 mg) are allowed, but dose reductions < 150 mg BID are not permitted.

SAFETY ANALYSES

- The safety analysis set included all randomly assigned patients who received ≥1 dose of any study treatment.
- AE and laboratory abnormality severity were graded using the National Cancer Institute Common Terminology Criteria for Adverse Events version 5.0 and classified by preferred term using the Medical Dictionary for Regulatory Activities.
- Treatment-emergent AEs (TEAEs), laboratory values, and TUC/PBO dose modifications and discontinuations were examined.

Figure 1. HER2CLIMB-05 study design

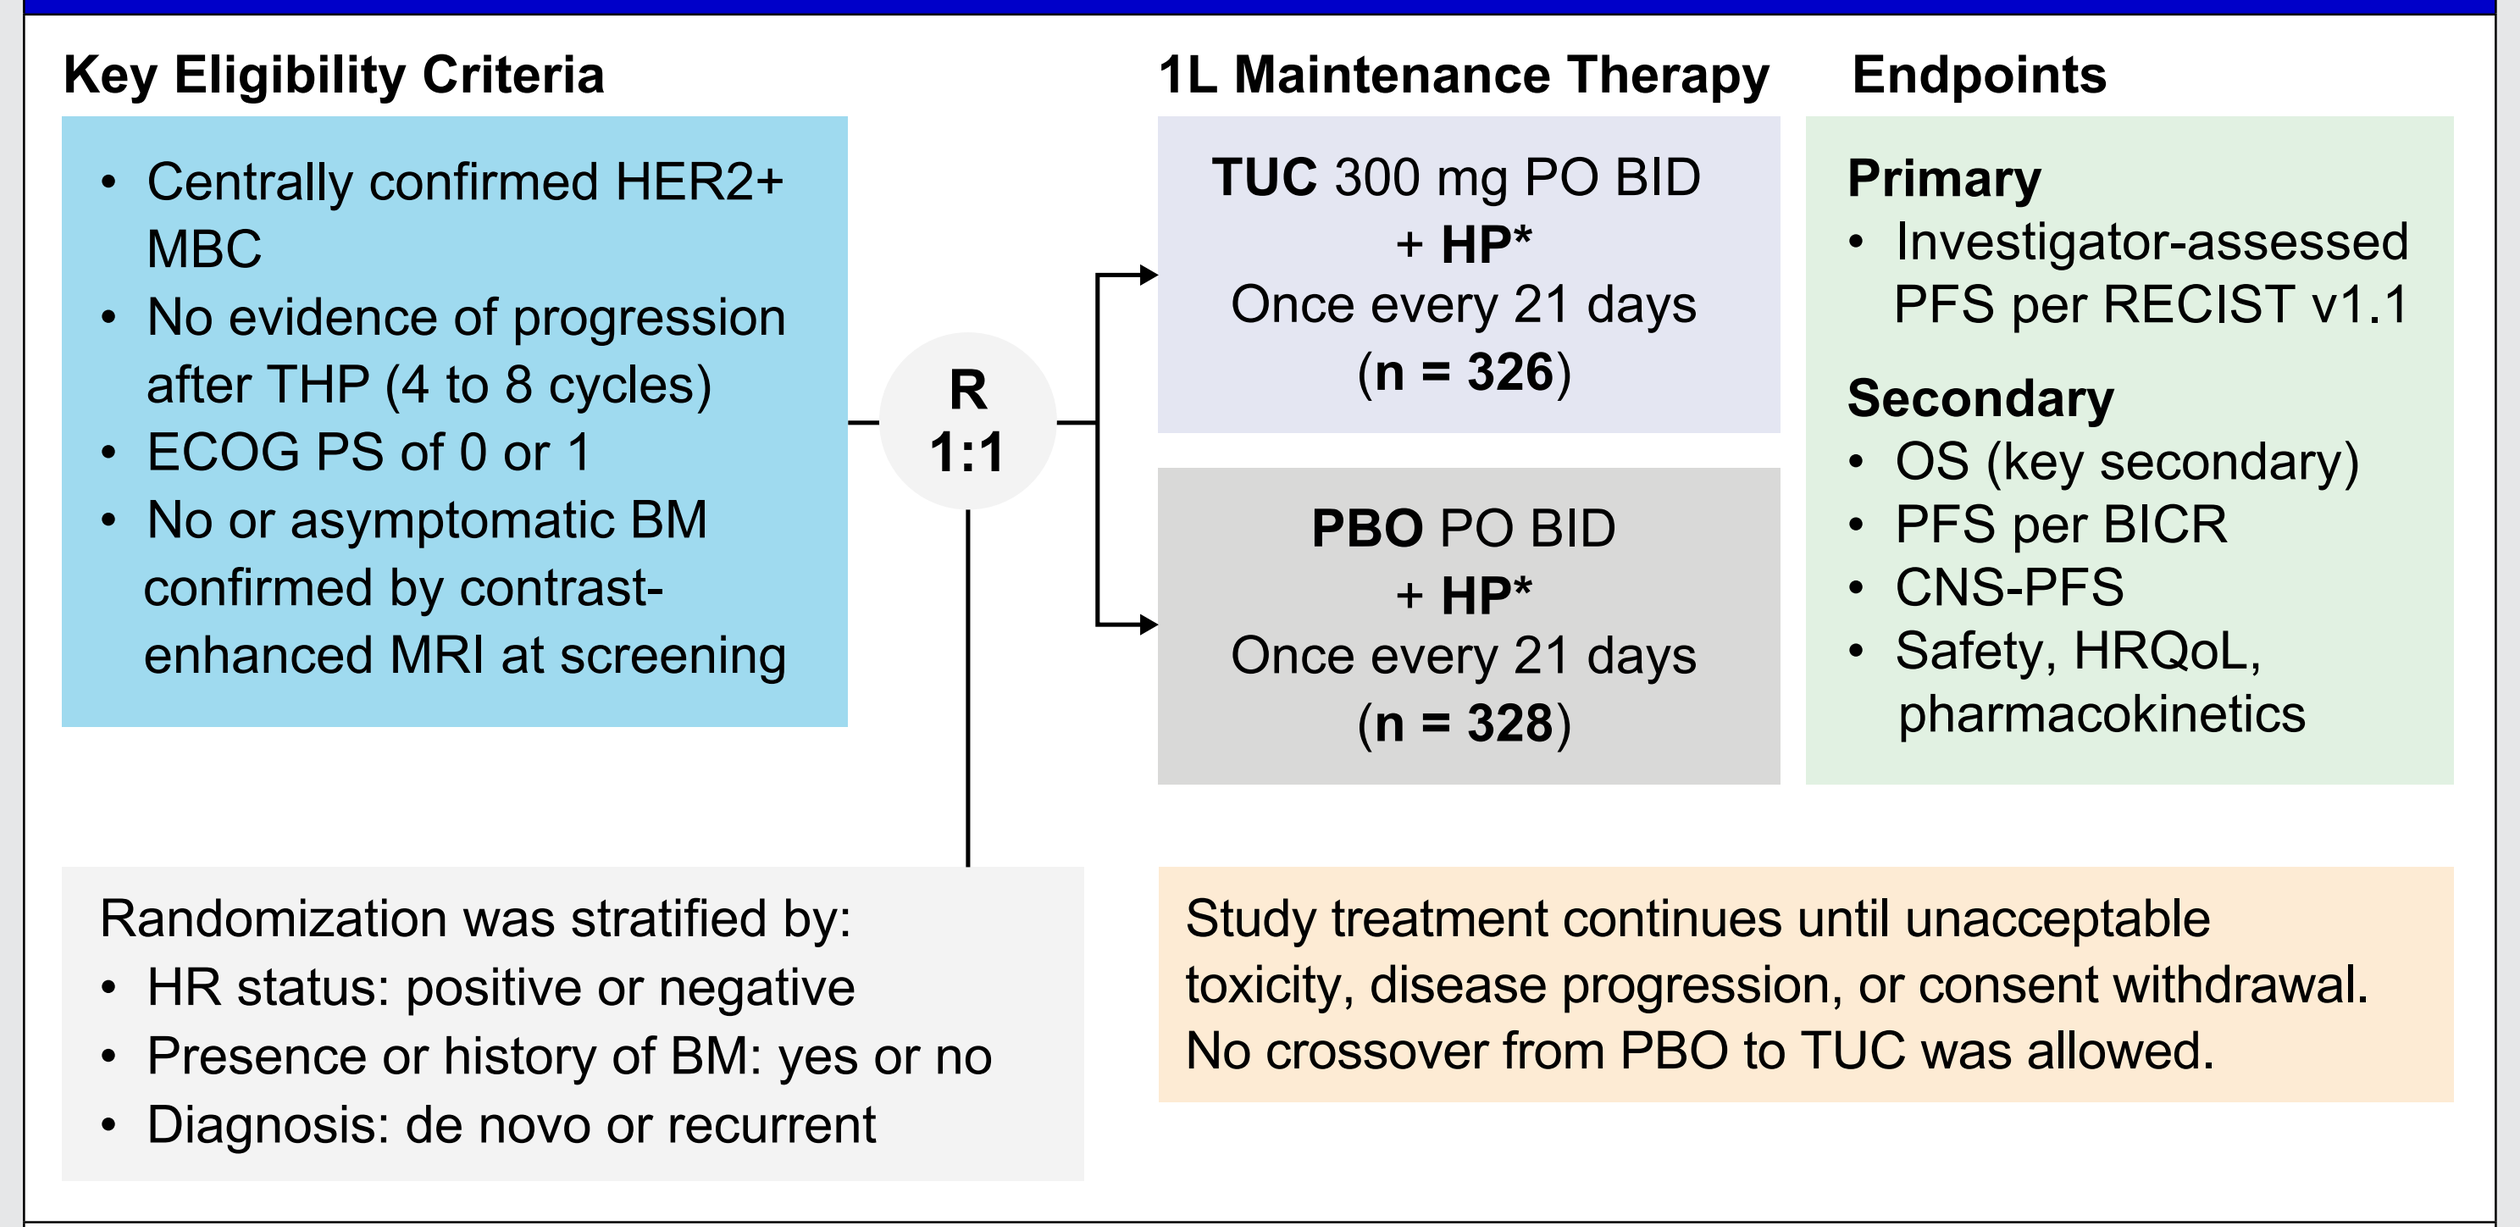
Key Eligibility Criteria
• Centrally confirmed HER2+ MBC
• No evidence of progression after THP (4 to 8 cycles)
• ECOG PS of 0 or 1
• No or asymptomatic BM confirmed by contrast-enhanced MRI at screening
R 1:1
1L Maintenance Therapy
TUC 300 mg PO BID + HP* Once every 21 days (n = 326)
PBO PO BID + HP* Once every 21 days (n = 328)
Endpoints
Primary
• Investigator-assessed PFS per RECIST v1.1
Secondary
• OS (key secondary)
• PFS per BICR
• CNS-PFS
• Safety, HRQoL, pharmacokinetics
Randomization was stratified by:
• HR status: positive or negative
• Presence or history of BM: yes or no
• Diagnosis: de novo or recurrent
Study treatment continues until unacceptable toxicity, disease progression, or consent withdrawal. No crossover from PBO to TUC was allowed.

*Concomitant ET was permitted during the study.
1L = first-line; BICR = blinded independent central review; BM = brain metastases; CNS = central nervous system; ECOG PS = Eastern Cooperative Oncology Group performance status; ET = endocrine therapy; H = trastuzumab; HER2+ = human epidermal growth factor receptor 2-positive; HR, hormone receptor; HRQoL = health-related quality of life; IV = intravenous; MBC, metastatic breast cancer; MRI = magnetic resonance imaging; OS = overall survival; P = pertuzumab; PBO = placebo; PFS = progression-free survival; PO BID = orally twice a day; R = randomization; RECIST = Response Evaluation Criteria in Solid Tumors; T = taxane; TUC = tucatinib.

Results

- Between March 2022 and July 2024, 654 patients were randomly assigned to the TUC arm (n = 326) or the PBO arm (n = 328).
- Of the randomly assigned patients, 4 never received any study treatment, 3 in the TUC arm and 1 in the PBO arm.[5] Additionally, 3 patients assigned to the PBO arm received ≥1 dose of TUC due to dispensing errors and were included in the TUC arm for the safety analyses.[5] Thus, the safety analysis set included 326 patients in the TUC arm and 324 in the PBO arm.
- At data cutoff (Sep 5, 2025), median TUC treatment duration was 17.1 months (range: 0.4–36.5). Treatment duration in the PBO arm was 15.5 months (range: 0.5–41.3).
- Most patients in the TUC and PBO arms experienced a TEAE; grade ≥3 TEAEs occurred more frequently in the TUC arm (**Table 1**).
- The most common TEAEs are shown in **Figure 2**.[5]

Table 1. Overall safety summary

Arm	TUC + HP (n = 326)	PBO + HP (n = 324)
Patients with TEAE, n (%)		
Any grade TEAEs	323 (99.1)	313 (96.6)
Grade ≥3 TEAEs	138 (42.3)	79 (24.4)
Serious TEAEs	55 (16.9)	26 (8.0)
TEAEs leading to death	1 (0.3)*	1 (0.3)

*A single grade 5 case of severe DILI was reported; however, the event was confounded by multiple concomitant medications with known hepatotoxic potential.
DILI = drug induced liver injury; H = trastuzumab; P = pertuzumab; PBO = placebo; TEAE = treatment-emergent adverse event; TUC = tucatinib.

Figure 2. TEAEs in HER2CLIMB-05 (≥20% of patients)

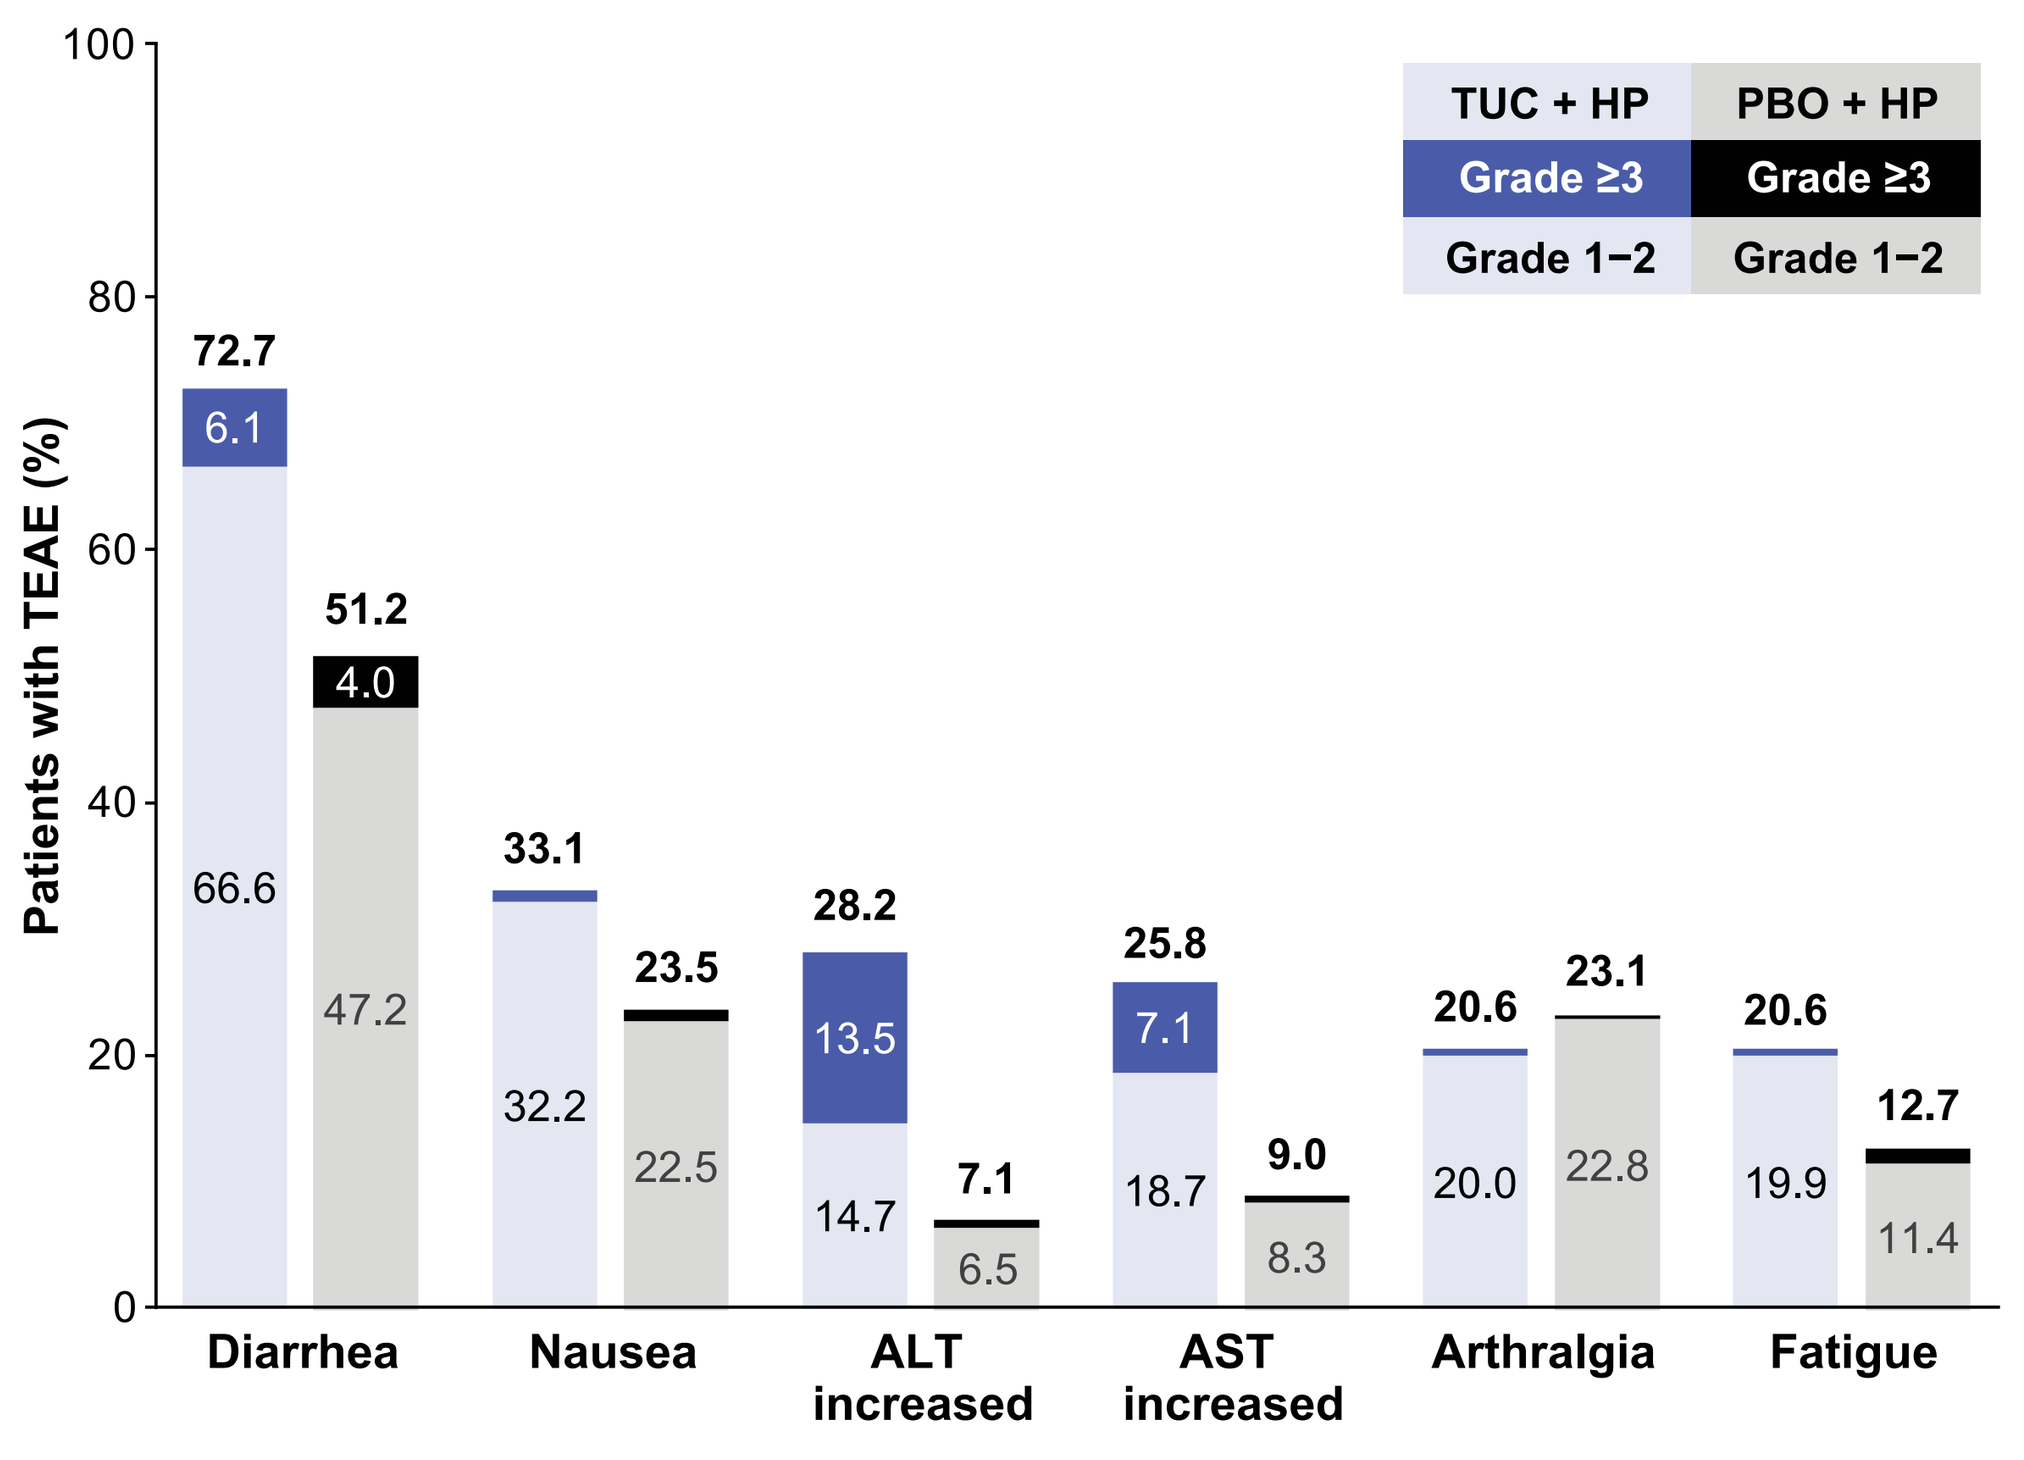
Patients with TEAE (%)
TUC + HP
PBO + HP
Grade ≥3
Grade 1–2
72.7
6.1
66.6
51.2
4.0
47.2
33.1
32.2
23.5
22.5
28.2
13.5
14.7
7.1
6.5
25.8
7.1
18.7
9.0
8.3
20.6
20.0
23.1
22.8
20.6
19.9
12.7
11.4
Diarrhea
Nausea
ALT increased
AST increased
Arthralgia
Fatigue

ALT = alanine aminotransferase; AST = aspartate aminotransferase; H = trastuzumab; P = pertuzumab; PBO = placebo; TEAE = treatment-emergent adverse event; TUC = tucatinib.

- Dose holds, reductions, and discontinuations due to TEAEs were more common in the TUC arm vs the PBO arm (**Figure 3**). However, relative dose intensity (median 95.4%, range: 16–119) remained high.
- Due to any grade TEAEs, 14.4% of patients had their TUC dose reduced by 1 level (250 mg BID); 8.3% reduced TUC dose by 2 levels (200 mg BID) and only 6.4% of the patients had their dose reduced to the lowest TUC dose (150 mg BID).
- TUC was discontinued in 13.5% patients due to TEAEs, mostly due to hepatic events.

Figure 3. Dose modifications/discontinuations due to any TEAEs

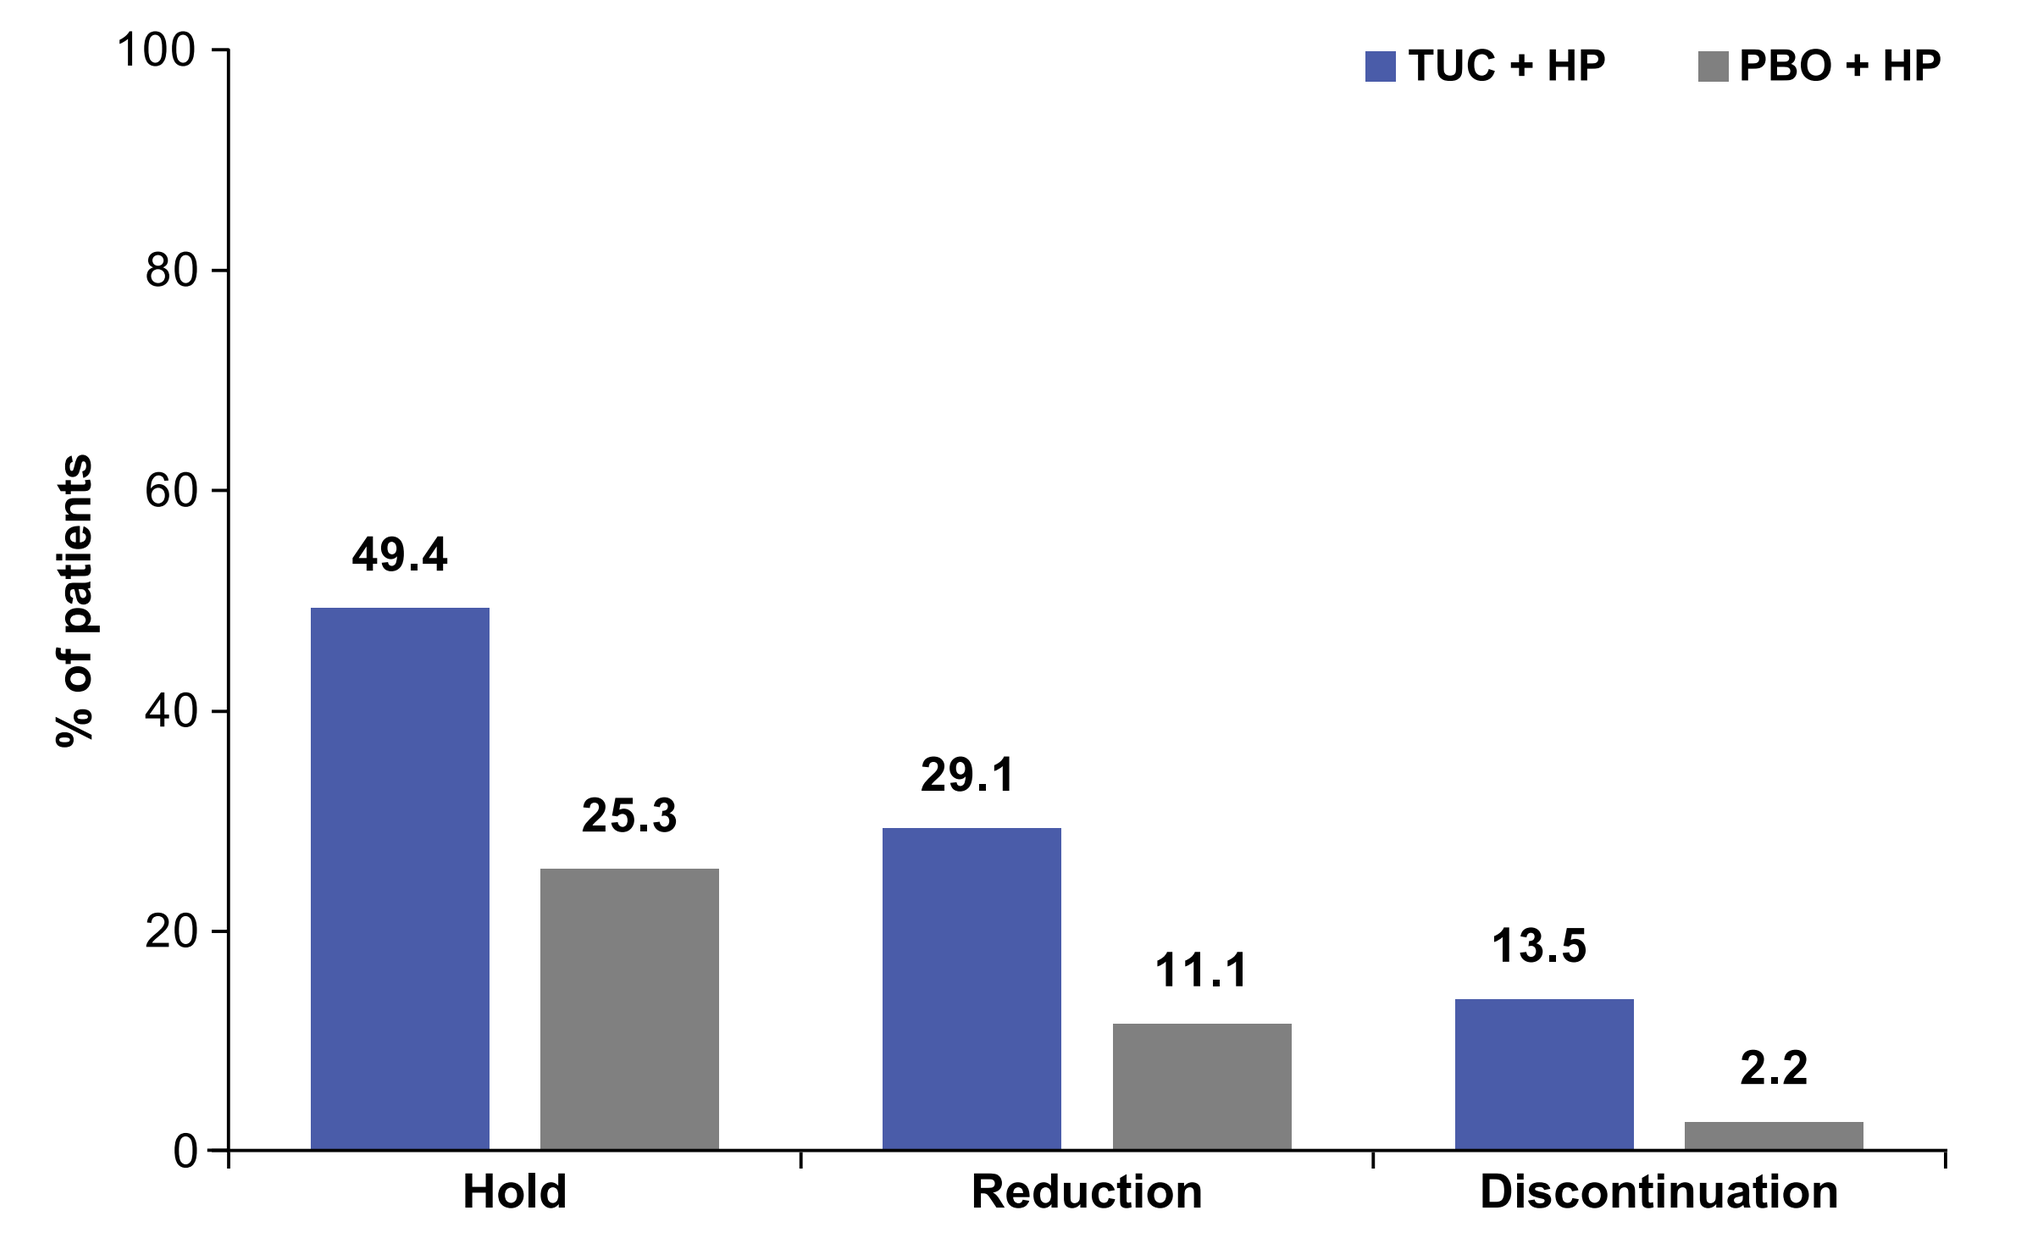
% of patients
TUC + HP
PBO + HP
49.4
25.3
29.1
11.1
13.5
2.2
Hold
Reduction
Discontinuation

H = trastuzumab; P = pertuzumab; PBO = placebo; TEAE = treatment-emergent adverse event; TUC = tucatinib.

Hepatic TEAEs

- Any grade and grade ≥3 hepatic TEAEs occurred at higher incidence in the TUC arm (**Table 2**). Increased ALT and AST accounted for the majority of these events, which were mostly asymptomatic, transient, and manageable with dose modifications and/or discontinuations, with no severe outcomes observed.
- Approximately half of the patients (n = 70) with a hepatic TEAE (any grade) were managed with TUC dose modifications or discontinuation, with most being attributed to elevation in ALT or AST (**Figure 4**).
- Due to a hepatic TEAE, 8.6% of patients had their TUC dose reduced by 1 level (250 mg BID); 4.6% reduced TUC dose by 2 levels (200 mg BID) and only 3.1% reduced TUC to the lowest dose (150 mg BID).
- 25 (7.7%) patients discontinued TUC due to hepatic TEAEs.

Diarrhea

- Any grade diarrhea occurred at higher incidence in the TUC arm vs the PBO arm, but most events in both treatment arms were of low severity and resolved quickly (**Table 3**).
- Grade 3 diarrhea occurred in 6.1% of patients in the TUC arm and no grade 4 TEAEs occurred. Serious diarrhea events were reported in 3 (0.9%) patients.
- Among the patients in the TUC arm, 62.9% used an antidiarrheal for a median duration of 5.1 days, while 40.1% in the PBO arm took an antidiarrheal for a median of 3.0 days. In either treatment arm, the most used medication was loperamide.
 - In the TUC arm, across all treatment cycles when a diarrhea event was reported, an antidiarrheal medicine was taken in 38.2% of them.

Table 2. Summary of hepatic TEAEs

Arm	TUC + HP (n = 326)		PBO + HP (n = 324)	
Hepatic TEAE, n (%)				
Any grade	142 (43.6)		51 (15.7)	
Grade ≥3	59 (18.1)		4 (1.2)	
Serious	13 (4.0)		1 (0.3)	
Hepatic TEAE reported by PT of investigators,* n (%)	**All grade**	**Grade ≥3**	**All grade**	**Grade ≥3**
ALT increased	92 (28.2)	44 (13.5)	23 (7.1)	2 (0.6)
AST increased	84 (25.8)	23 (7.1)	29 (9.0)	2 (0.6)
Blood bilirubin increased	20 (6.1)	3 (0.9)	10 (3.1)	0
GGT increased	12 (3.7)	2 (0.6)	5 (1.5)	1 (0.3)
Hepatic function abnormal	11 (3.4)	4 (1.2)	3 (0.9)	0
Hepatic cytolysis	9 (2.8)	3 (0.9)	3 (0.9)	0
Hypertransaminasemia	7 (2.1)	3 (0.9)	1 (0.3)	0
Drug-induced liver injury	6 (1.8)	3 (0.9)	0	0
Liver injury	4 (1.2)	3 (0.9)	1 (0.3)	0
Median time, days (range)				
To onset of first event	34.5 (4–776)	40.0 (4–492)	85.0 (6–826)	386.5 (77–610)
To resolution	25.0 (2–328)	40.0 (5–217)	22.0 (2–302)	10.0 (7–96)

*Occurring in >1% of patients in the TUC arm.
ALT = alanine aminotransferase; AST = aspartate aminotransferase; GGT = gamma-glutamyl transferase; H = trastuzumab; P = pertuzumab; PBO = placebo; PT = preferred term; TEAE = treatment-emergent adverse event; TUC = tucatinib.

Figure 4. Dose modifications/discontinuations due to hepatic TEAEs

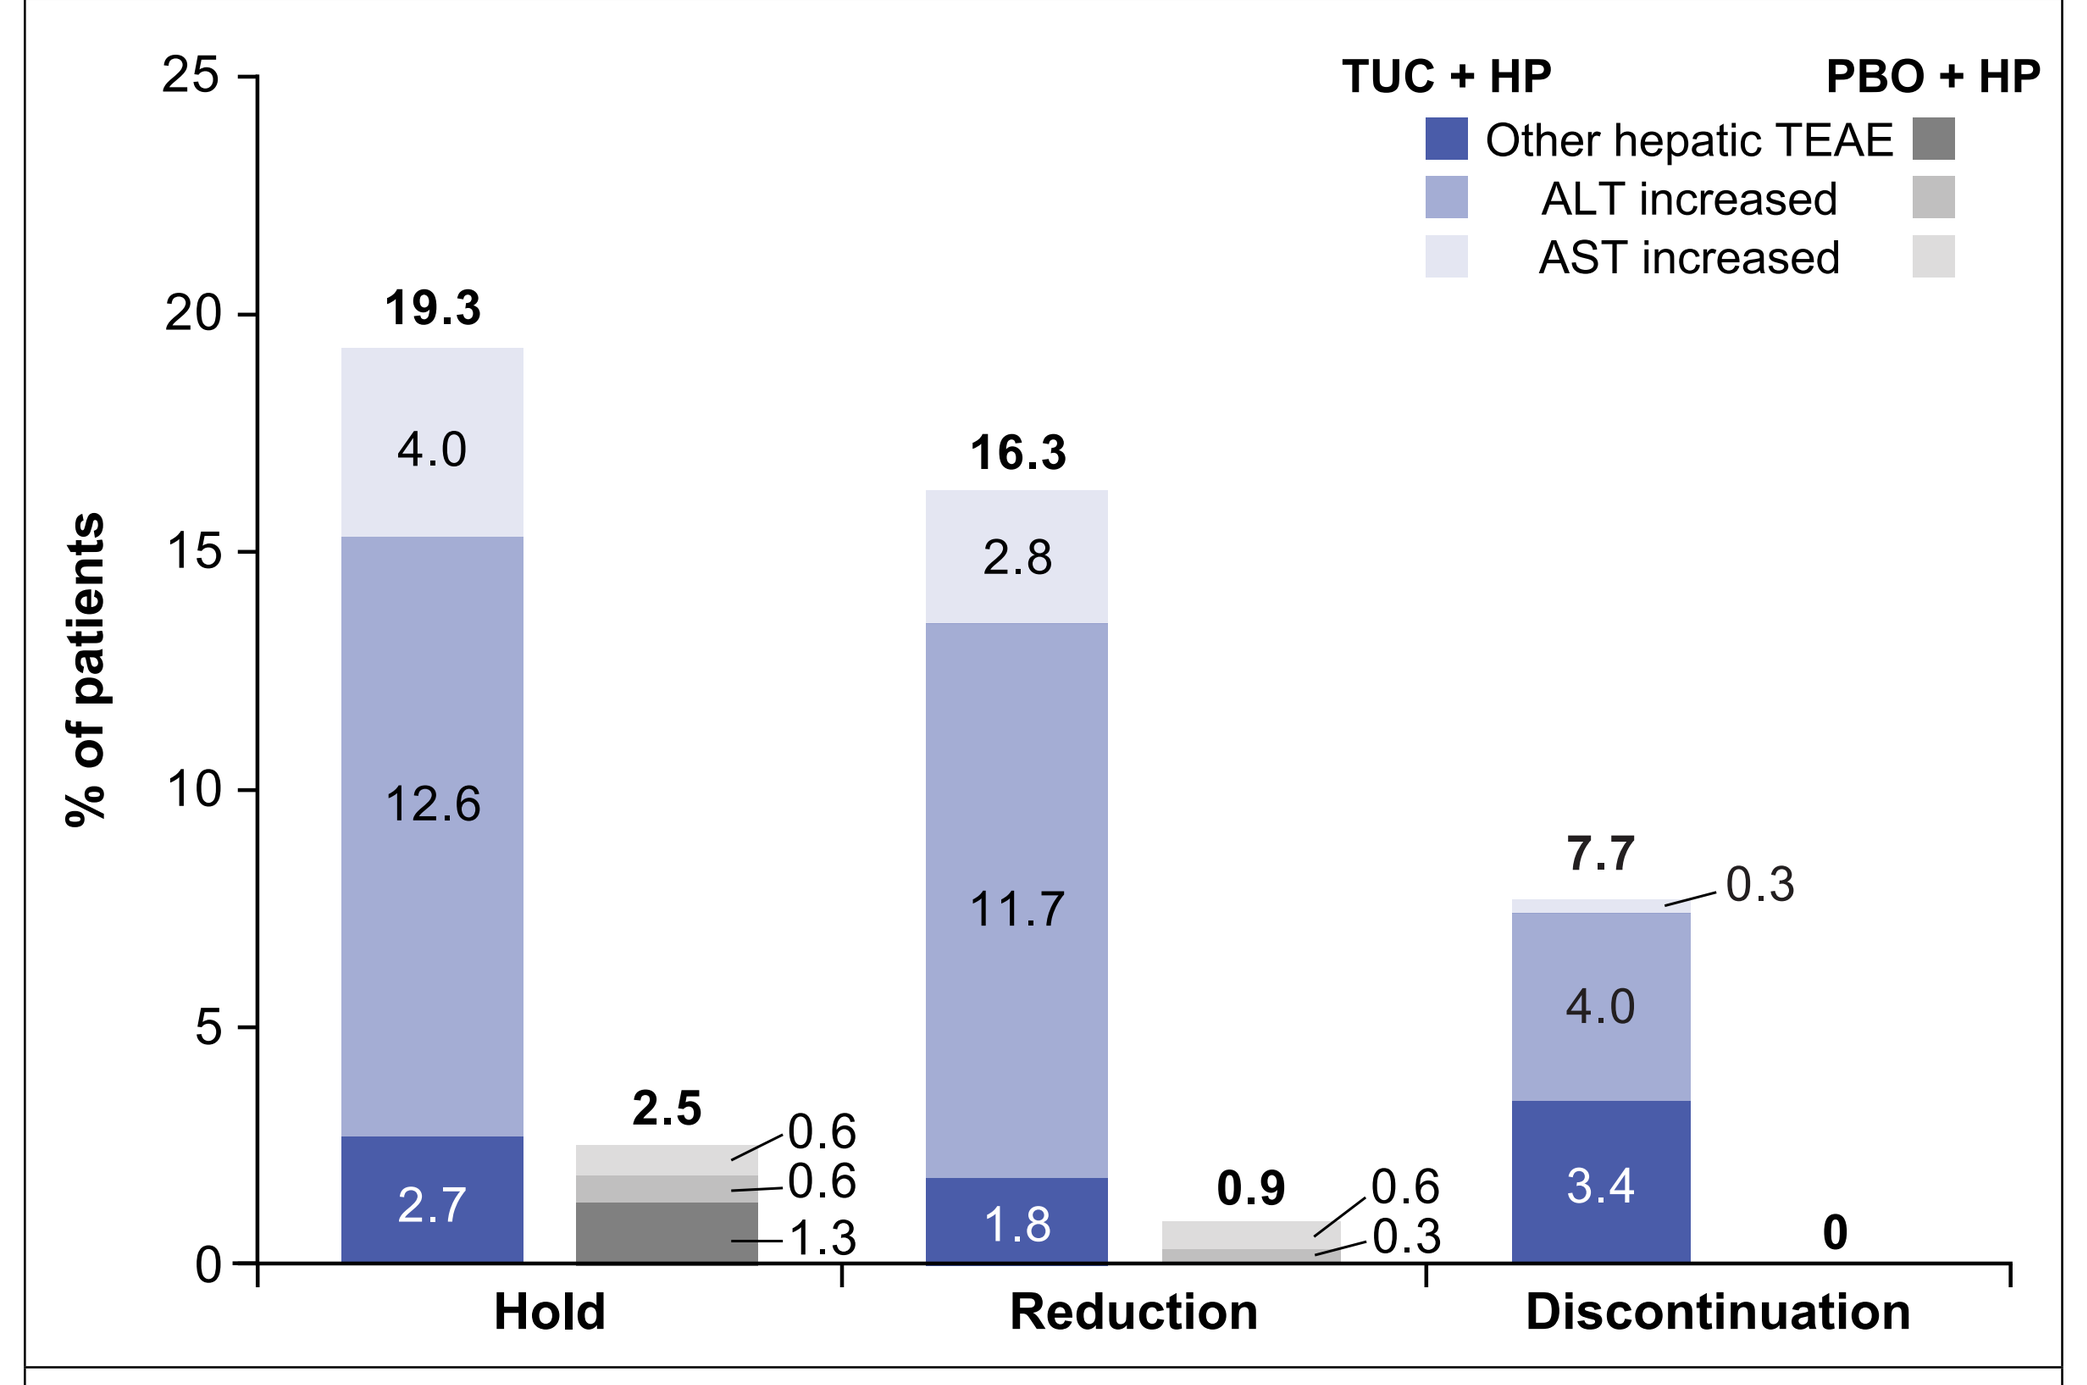
% of patients
TUC + HP
PBO + HP
Other hepatic TEAE
ALT increased
AST increased
19.3
4.0
12.6
2.7
2.5
0.6
0.6
1.3
16.3
2.8
11.7
1.8
0.9
0.6
0.3
7.7
0.3
4.0
3.4
0
Hold
Reduction
Discontinuation

ALT = alanine aminotransferase; AST = aspartate aminotransferase; H = trastuzumab; P = pertuzumab; PBO = placebo; TEAE = treatment-emergent adverse event; TUC = tucatinib.

Table 3. Summary of diarrhea TEAEs

Arm	TUC + HP (n = 326) All grades	TUC + HP Grade 2	TUC + HP Grade 3	PBO + HP (n = 324) All grades	PBO + HP Grade 2	PBO + HP Grade 3
Diarrhea TEAE, n (%)	237 (72.7)	103 (31.6)	20 (6.1)	166 (51.2)	43 (13.3)	13 (4.0)
Median time, days (range)						
To onset of first event	11.0 (1–683)	32.0 (1–736)	49.5 (2–350)	32.0 (1–768)	104.0 (2–506)	149.0 (6–626)
To resolution	2.0 (1–760)	5.0 (1–585)	8.0 (1–162)	5.0 (1–585)	4.0 (1–578)	14.0 (1–309)
Antidiarrheal use for any grade diarrhea, n (%)	205 (62.9)			130 (40.1)		
Median duration, days	5.1 (1–21)			3.0 (1–23)		

H = trastuzumab; P = pertuzumab; PBO = placebo; TEAE = treatment-emergent adverse event; TUC = tucatinib.

- Only 11.3% of patients in the TUC arm were managed with dose modifications and/or discontinuations due to diarrhea (**Figure 5**).
- Due to diarrhea, 2.5% of patients had their TUC dose reduced by 1 level (250 mg BID); 1.8% reduced TUC dose by 2 levels (200 mg BID) and only 2.1% reduced TUC to the lowest dose (150 mg BID).
- Per study protocol, TUC dose modification is only recommended for grade ≥3 diarrhea. However, among the patients with maximum severity of grade 2 diarrhea (n = 103) in the TUC arm, 8.7% had a TUC dose hold, 6.8% had TUC dose reduced, and 1 patient discontinued TUC.

Figure 5. Dose modifications/discontinuations due to diarrhea

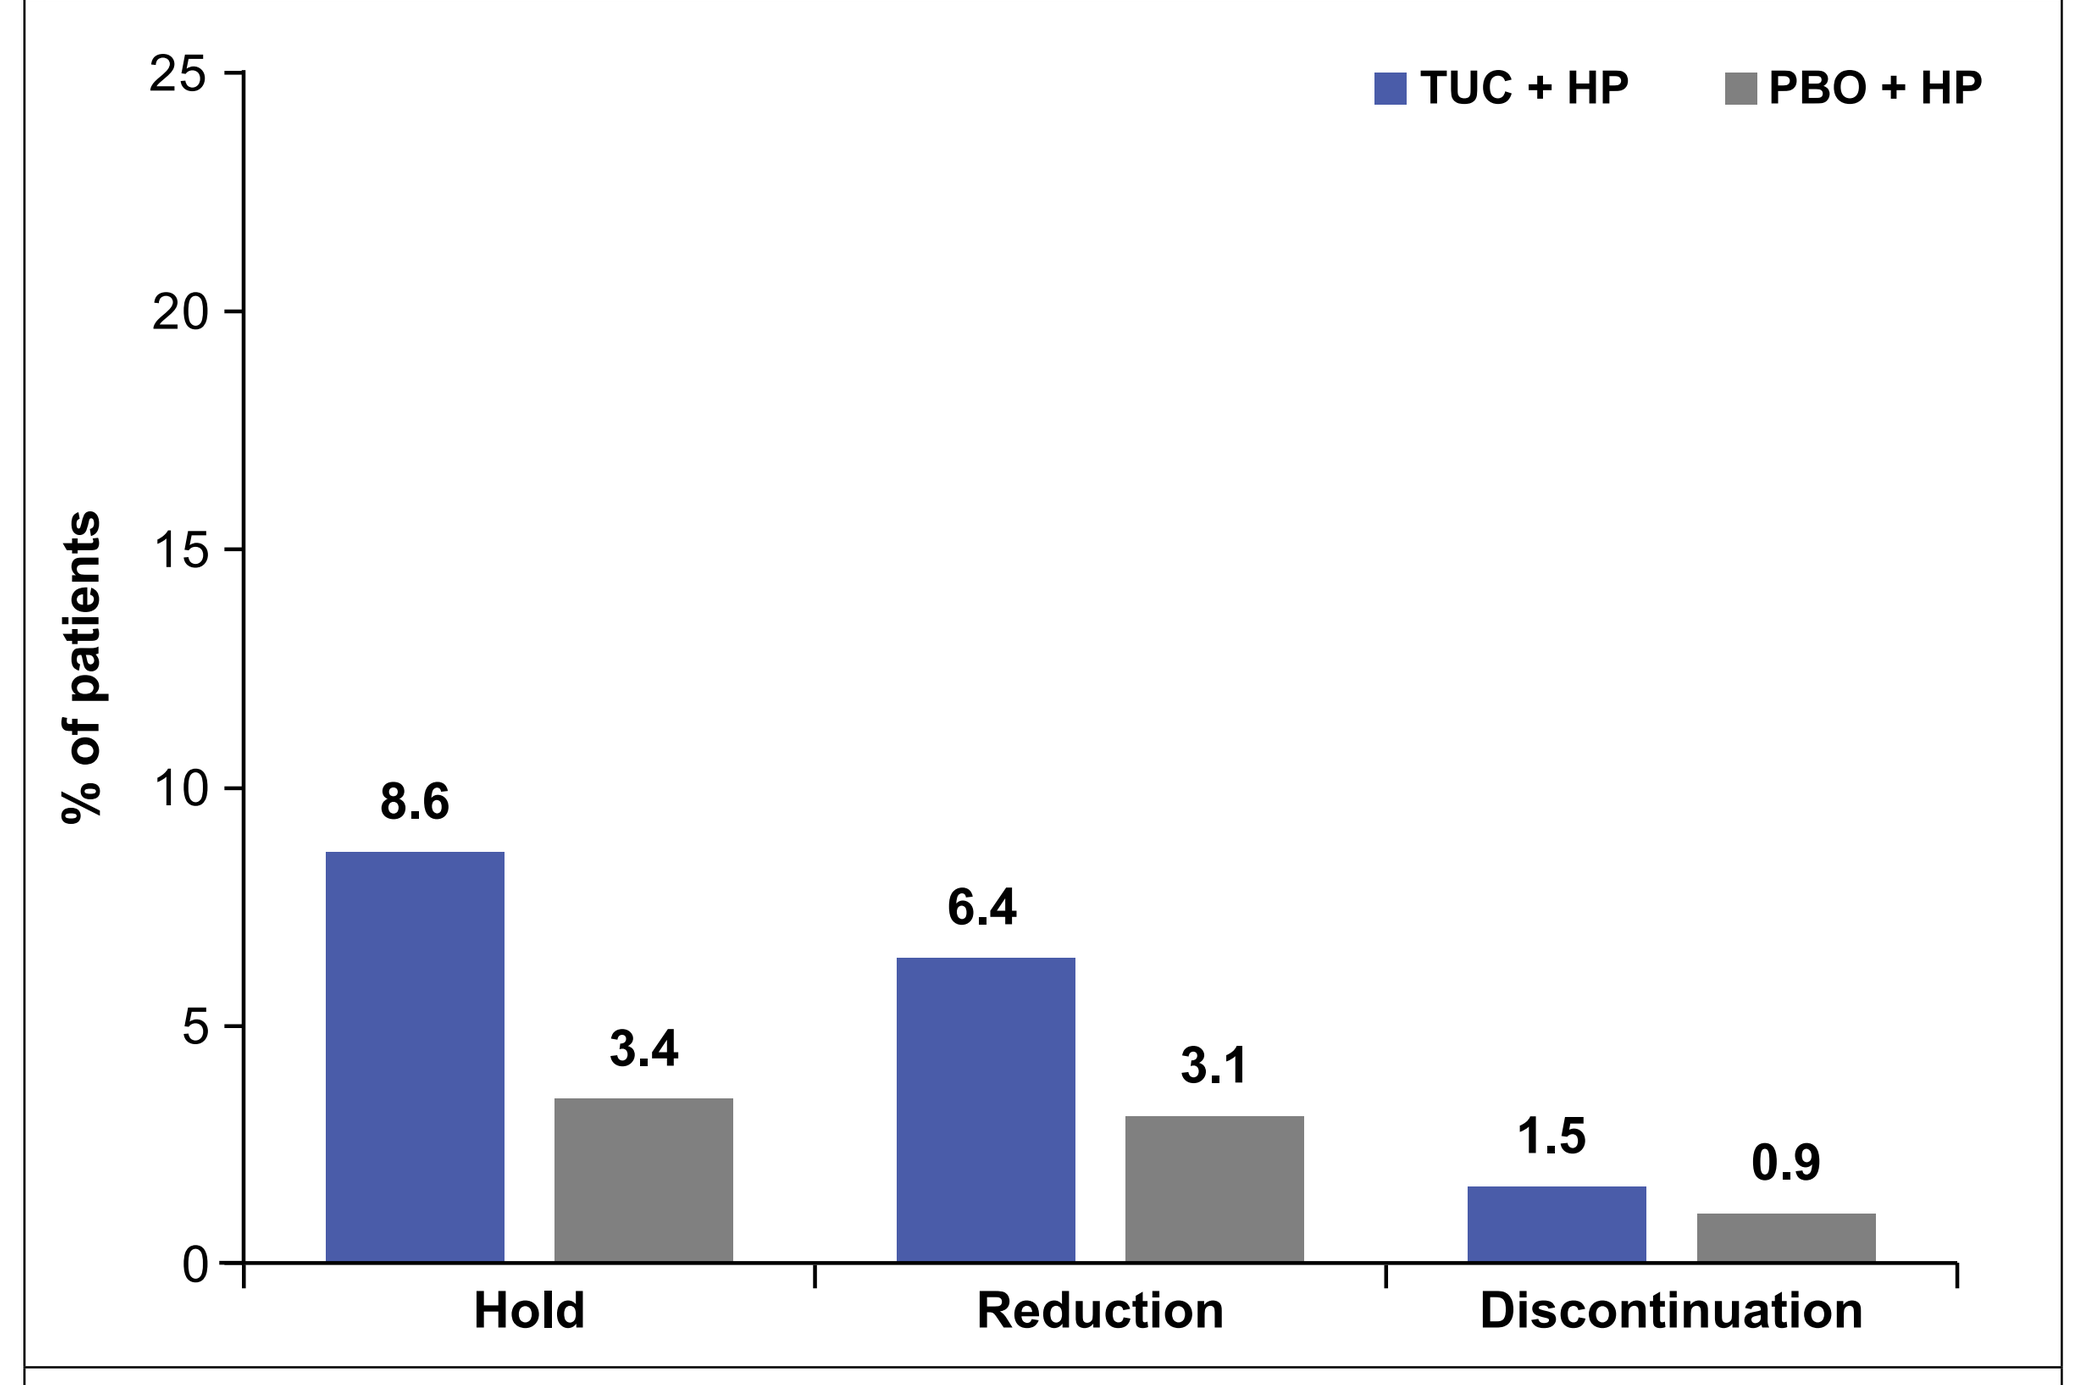
% of patients
TUC + HP
PBO + HP
8.6
3.4
6.4
3.1
1.5
0.9
Hold
Reduction
Discontinuation

H = trastuzumab; P = pertuzumab; PBO = placebo; TUC = tucatinib.

Copyright © 2026. All rights reserved.