751P

Subsequent therapies in the phase 3 BREAKWATER study of first-line encorafenib + cetuximab + mFOLFOX6 in patients with BRAF V600E-mutant metastatic colorectal cancer

Josep Tabernero,[1] Eric Van Cutsem,[2] Cathy Eng,[3] Takayuki Yoshino,[4] Harpreet Wasan,[5] Fortunato Ciardiello,[6] Timothy Maughan,[7,8] Tae Won Kim,[9] Rona Yaeger,[10] Jayesh Desai,[11] Elena Élez,[1] Sara Lonardi,[12] Lin Shen,[13] Ying Yuan,[14] Van Morris,[15] Tiziana Usari,[16] Katherine Liau,[17] Kelechi Olu,[18] Xiaosong Zhang,[19] Scott Kopetz[15]

[1]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, UVic-UCC, Barcelona, Spain; [2]University Hospital Gasthuisberg and University of Leuven, Leuven, Belgium; [3]Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [4]National Cancer Center Hospital East, Kashiwa, Japan; [5]Hammersmith Hospital, Division of Cancer, Imperial College, London, UK; [6]University of Campania Luigi Vanvitelli, Naples, Italy; [7]University of Liverpool, Liverpool, UK; [8]MRC Oxford Institute for Radiation Oncology, University of Oxford, Oxford, UK; [9]Asan Medical Center, University of Ulsan College of Medicine, Seoul, South Korea; [10]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [11]Peter MacCallum Cancer Centre, Melbourne, Australia; [12]Veneto Institute of Oncology-IRCCS, Padova, Italy; [13]Peking University Cancer Hospital and Institute, Beijing, China; [14]The second Affiliated Hospital of College of Medicine, Zhejiang University, Hangzhou, China; [15]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [16]Pfizer, Inc, Milan, Italy; [17]Pfizer, Inc, La Jolla, CA, USA; [18]Pfizer AG, Switzerland; [19]Pfizer, Inc, South San Francisco, CA, USA

Objectives

- We present data on subsequent therapies received by patients enrolled in the phase 3 BREAKWATER study of 1L EC ± mFOLFOX6 vs control in BRAF V600E-mutant mCRC

Conclusions

- The majority of patients who discontinued study treatment in BREAKWATER received subsequent systemic anticancer therapy
- Of the patients in the control arm who received subsequent therapies, the majority received BRAF inhibitor–based targeted therapies, most frequently in the 2L
- This treatment pattern corresponds to current real-world clinical practice and supports that the improved outcomes observed with EC+mFOLFOX6 in the 1L are based on evaluation against valid current treatments
- PFS after subsequent therapy (PFS2) was longer in the EC+mFOLFOX6 arm than in patients in the control arm who received EC-based treatment in the 2L, suggesting a long-term clinical benefit of upfront treatment with EC+mFOLFOX6 vs sequential use of 1L chemotherapy and 2L EC
- Together with the statistically significant and clinically meaningful improvement in OS with EC+mFOLFOX6 vs control, these results further support EC+mFOLFOX6 as a current 1L standard of care for BRAF V600E-mutant mCRC

Plain Language Summary
Please scan this quick response (QR) code with your smartphone to view a plain language summary. If you do not have a smartphone, access the plain language summary via the internet at: https://scientificpubs.congressposter.com/pls/tnxtnwczmqgbvbzg

Electronic Poster
Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors. If you do not have a smartphone, access the poster via the internet at:
https://scientificpubs.congressposter.com/p/tnxtnwczmqgbvbzg

References: **1.** Tabernero J, et al. *Am Soc Clin Oncol Educ Book*. 2022;42:254-63. **2.** Tran B, et al. *Cancer*. 2011;117:4623-32. **3.** Delord JP, et al. *Clin Cancer Res*. 2017;23:5339-48. **4.** Stuart DD, et al. *Cancer Res*. 2012;72(8_Suppl):3790. **5.** Morris VK, et al. *J Clin Oncol*. 2023;41:678-700. 6. Cohen R, et al. *J Natl Cancer Inst*. 2021;113:1386-95. **7.** Cremolini C, et al. *J Clin Oncol*. 2020;38:3314-24. **8.** Kopetz et al, *Nat Med*. 2025;31:901–8. **9.** Élez E, et al. *N Engl J Med*. 2025; 392:2425-2437. **10.** BRAFTOVI Prescribing information 2024. US Food and Drug Administration.

Abbreviations: 1L, first line; 2L, second line; 3L, third line; 4L, fourth line; 5L, fifth line; 6L, sixth line; 7L, seventh line; ATP, adenosine triphosphate; BICR, blinded independent central review; CAPOX, capecitabine/oxaliplatin; DOR, duration of response; dMMR, deficient mismatch repair; EC, encorafenib+cetuximab; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; FDA, Food and Drug Administration; FOLFOXIRI, fluorouracil/leucovorin/oxaliplatin/irinotecan; HR, hazard ratio; mCRC, metastatic colorectal cancer; mFOLFOX6, modified fluorouracil/leucovorin/oxaliplatin; MSI-H, microsatellite instability-high; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PFS2, progression-free survival 2; PRO, patient reported outcomes; R, randomized; RECIST, Response Evaluation Criteria in Solid Tumors; TTR, time to response.

Acknowledgments: The authors thank the participating patients and their families, as well as the staff at the participating sites.

BREAKWATER was sponsored by Pfizer and was conducted with support from ONO Pharmaceutical, Merck KGaA, Darmstadt, Germany, and Eli Lilly and Company.

Medical writing support was provided by Tina Wasmeier, PhD, of Nucleus Global, an Inizio Company, and was funded by Pfizer.

Disclosures/COIs: **Josep Tabernero** has a consulting or advisory role at Array BioPharma, AstraZeneca, Bayer, Boehringer Ingelheim, Chugai Pharma, Daiichi Sankyo, F. Hoffmann-La Roche, Genentech, HalioDx, Hutchison MediPharma, Ikena Oncology, Inspirna, IQVIA, Lilly, Menarini, Merck Serono, Merus, Mirati Therapeutics, MSD, NeoPhore, Novartis, Ona Therapeutics, Orion Biotechnology, Peptomyc, Pfizer, Pierre Fabre, Samsung Bioepis, Sanofi, Scandion Oncology, Scorpion Therapeutics, Seagen, Servier, Taiho Pharmaceutical, Tessa Therapeutics and TheraMyc and has other relationships with Amgen, Array Biopharma, BeiGene, Boehringer Ingelheim, BMS, Cancer Research UK, Celgene, Debiopharm, F. Hoffman-La Roche, Fundación Científica de la Asociación Española Contra el Cáncer, Genentech, HalioDx, Hutchison Medipharma, Imedex, Janssen-Cilag, MedImmune, Medscape, Menarini, Merck Health KGaA, MJH Life Sciences, MSD, Merus, Mirati, Novartis, Oniria Therapeutics, PeerView Institute for Medical Education, Pfizer, PharmaMar, Physicans' Education Resource, Sanofi-Aventis, Servier and Taiho Pharmaceutical. **Eric Van Cutsem** has a consulting or advisory role at AbbVie, Agenus, ALX, Amgen, Arcus Biosciences, Astellas Pharma, AstraZeneca, Bayer, BeiGene, BioNTech, Boehringer Ingelheim, Bristol Myers Squibb, Daiichi Sankyo, Debiopharm, ElmediX, Eisai, GSK, Hookipa Biotech, Incyte, Ipsen, Lilly, Merck Sharp & Dohme, Merck KGaA, Mirati, Novartis, Nordic, Pierre Fabre, Pfizer, Roche, Seagen, Servier, Simcere, Takeda, Taiho Pharmaceutical and Terumo. **Cathy Eng** has a consulting role at Bayer, Boston Scientific, GlaxoSmithKline, HalioDx, Merck, Mirati, Hookipa, J&J, Natera, Roche, Seagen, Taiho and Veloxis and receives research funding (to VUMC) from Elevar, Hutchinson, Merck and Pfizer. **Takayuki Yoshino** receives honoraria from Chugai Pharma, Merck KGaA, MSD, Ono Pharmaceutical, and Takeda, consulting fee from Indivumed and Suminoto Corp, and research funding from Amgen, Bristol-Mayers Squibb, Caris, Chugai Pharma, Daiichi Sankyo, Exact Sciences, Eisai, FALCO Biosystems, Medical & Biological Laboratories, Merus N.V., Miyarisan Pharmaceutical, Molecular Health, MSD, Natera, Nippon Boehringer Ingelheim, Ono Pharmaceutical, Pfizer, Roche Diagnostics, Sanofi, Sysmex, Takeda Pharmaceutical and Taiho Pharmaceutical. **Harpreet Wasan** has a consulting or advisory role at Amgen, Bayer, Bristol Myers Squibb (Celgene), Boehringer Ingelheim (DMC), BTG, EXACT Therapeutics, Erytech Pharma, Incyte, Merck KGaA, Oaktree Life Sciences, OncoSil, Pfizer, Pierre Fabre, Roche/Genentech, Seagen, Servier, Shire, Sirtex Medical, Takeda (Hutchinson Med) and Zymeworks and receives research funding (institutional) from Merck KGaA, MSD, Pfizer and Sirtex Medical. **Fortunato Ciardiello** has a consulting or advisory role at Amgen, Bayer, Merck KGaA, Pfizer and Roche/Genentech and receives research funding from Amgen, Bayer, Bristol Myers Squibb, Ipsen, Merck KGaA, MSD, Roche/Genentech, Servier and Symphogen. **Timothy Maughan** has a consulting or advisory role at AstraZeneca, Pierre Fabre and Vertex; receives research funding (institutional) from Almac Diagnostics, AstraZeneca, Merck KgaA and PsiOxus Therapeutics; and has a patent pending. **Tae Won Kim** .receives research funding (institutional) from AstraZeneca, Sanofi and Roche/ Genentech. **Rona Yaeger** has a consulting or advisory role at Mirati Therapeutics, Revolution Medicines, Eli Lilly, Merck, and Parabilis and receives research funding from Boundless Bio, Boehringer Ingelheim, Mirati Therapeutics, Pfizer, Parabilis and Daiichi Sankyo. **Jayesh Desai** has a consulting or advisory role at Amgen, Bayer, BeiGene, Daiichi Sankyo, Eisai, GSK, Merck KGaA and Pierre Fabre and receives research funding from AstraZeneca/MedImmune, BeiGene, Bionomics, Bristol Myers Squibb, GSK, Lilly, Novartis and Roche. **Elena Élez** has a consulting or advisory role at Agenus, Amgen, Bayer, Boehringer Ingelheim, Cureteq, F. Hoffmann-La Roche, GlaxoSmithKline, Janssen, Merck, MSD, Nordic Group BV, NovartisPierre Fabre, Repare Therapeutics, RIN Institute Inc., Rottapharm Biotech, Sanofi Aventis, Seagen, Servier and Takeda and has received speaker honoraria from Bristol-Myers Squibb Company, Lilly, Medscape, Organon & Co. and Pfizer. **Sara Lonardi** reports consulting or advisory role for Amgen, Lilly, Merck Serono and SERVIER; speakers' bureau from Bristol-Myers Squibb, Lilly, Merck Serono, Roche and Servier; research funding from Amgen and Merck Serono. Lin Shen reports research funding from Beigene and Pfizer (Inst). **Ying Yuan** reports research funding from Pfizer (Inst). **Van Morris** has a consulting or advisory role at Bristol-Myers Squibb, GlaxoSmithKline, Incyte Corporation, Scandion Oncology, reports research funding from Bristol-Myers Squibb and Pfizer and is an employee of The University of Texas MD Anderson Cancer Center. **Tiziana Usari** is an employee of and has stock and other ownership interests in Pfizer. **Katherine Liau** is an employee of and has stock and other ownership interests in Pfizer. **Kelechi Olu** is an employee of and has stock and other ownership interests in Pfizer. **Xiaosong Zhang** is an employee of and has stock and other ownership interests in Pfizer and has patents, royalties or other intellectual property via Johns Hopkins University. **Scott Kopetz** has stock and other ownership interests in Lylon, Lutris, MolecularMatch and Navire; a consulting or advisory role at AbbVie, Amal Therapeutics, AstraZeneca/MedImmune, Bayer Health, Bicara Therapeutics, Boehringer Ingelheim, Boston Biomedical, Carina Biotech, Daiichi Sankyo, EMD Serono, Endeavor BioMedicines, Flame Biosciences, Genentech, Gilead Sciences, GSK, HalioDx, Holy Stone Healthcare, Invivata, Ipsen, Iylon, Jacobio, Jazz Pharmaceuticals, Lilly, Lutris, Merck, Mirati Therapeutics, Natera, Novartis, Numab, Pfizer, Pierre Fabre, Redx Pharma, Repare Therapeutics, Servier and Xilis and received research funding from Amgen, Array BioPharma, Biocartis, Daiichi Sankyo, EMD Serono, Genentech/Roche, Guardant Health, Lilly, MedImmune, Novartis and Sanofi.

Presented at ESMO 2025; Oct 17-21, 2025; Berlin, Germany.

Background

- *BRAF* V600E mutations are present in 8% to 12% of patients with mCRC and are associated with poorer prognosis and reduced efficacy of standard chemotherapy compared with *BRAF* wild-type disease[1,2]
- Encorafenib is a highly selective, ATP-competitive BRAF inhibitor with longer pharmacodynamic activity than other approved BRAF inhibitors[3,4]
- Encorafenib in combination with the anti-EGFR monoclonal antibody cetuximab is established as standard care for previously treated BRAF V600E-mutated mCRC[5]
- In the 1L setting, treatment for BRAF V600E-mutated mCRC has historically consisted of chemotherapy with or without a biologic agent (eg, bevacizumab); however, efficacy is limited[6,7]
- The ongoing phase 3 BREAKWATER study (NCT04607421) is evaluating EC±mFOLFOX6 for 1L treatment of BRAF V600E-mutant mCRC[8,9]
- EC+mFOLFOX6 demonstrated statistically significant and clinically meaningful improvements in ORR, PFS, and OS vs chemotherapy with or without bevacizumab as control in BREAKWATER[8,9]
 - ORR was 60.9% vs 40.0% (odds ratio 2.443; *P*=0.0008),[8] median PFS was 12.8 vs 7.1 months (HR 0.53; *P*<0.001), and median OS was 30.3 vs 15.1 months (HR 0.49; *P*<0.001)
- EC+mFOLFOX6 was granted accelerated approval by the US FDA as part of Project FrontRunner for patients with BRAF V600E-mutant mCRC, including in the 1L setting[10]; this was followed by several recent full approvals in countries in Latin America and conditional approval in Canada
- EC+mFOLFOX6 is a new standard of care for 1L treatment of BRAF V600E-mutant mCRC

Methods

- BREAKWATER is an open-label, global, randomized, phase 3 study in patients with previously untreated BRAF V600E-mutant mCRC[8,9] (**Figure 1**)
- Patients were randomized 1:1:1 to receive EC, EC+mFOLFOX6, or control (investigator's choice of chemotherapy with or without bevacizumab)
- Subsequent anticancer therapies after discontinuing study treatment were per investigator discretion
- PFS2 was defined as the time from randomization to second objective disease progression (progressive disease after the start of subsequent anticancer therapy) per RECIST 1.1 (by investigator assessment), discontinuation of next-line treatment after first objective disease progression (by investigator assessment), or death from any cause, whichever occurs first

Figure 1. BREAKWATER Study Design

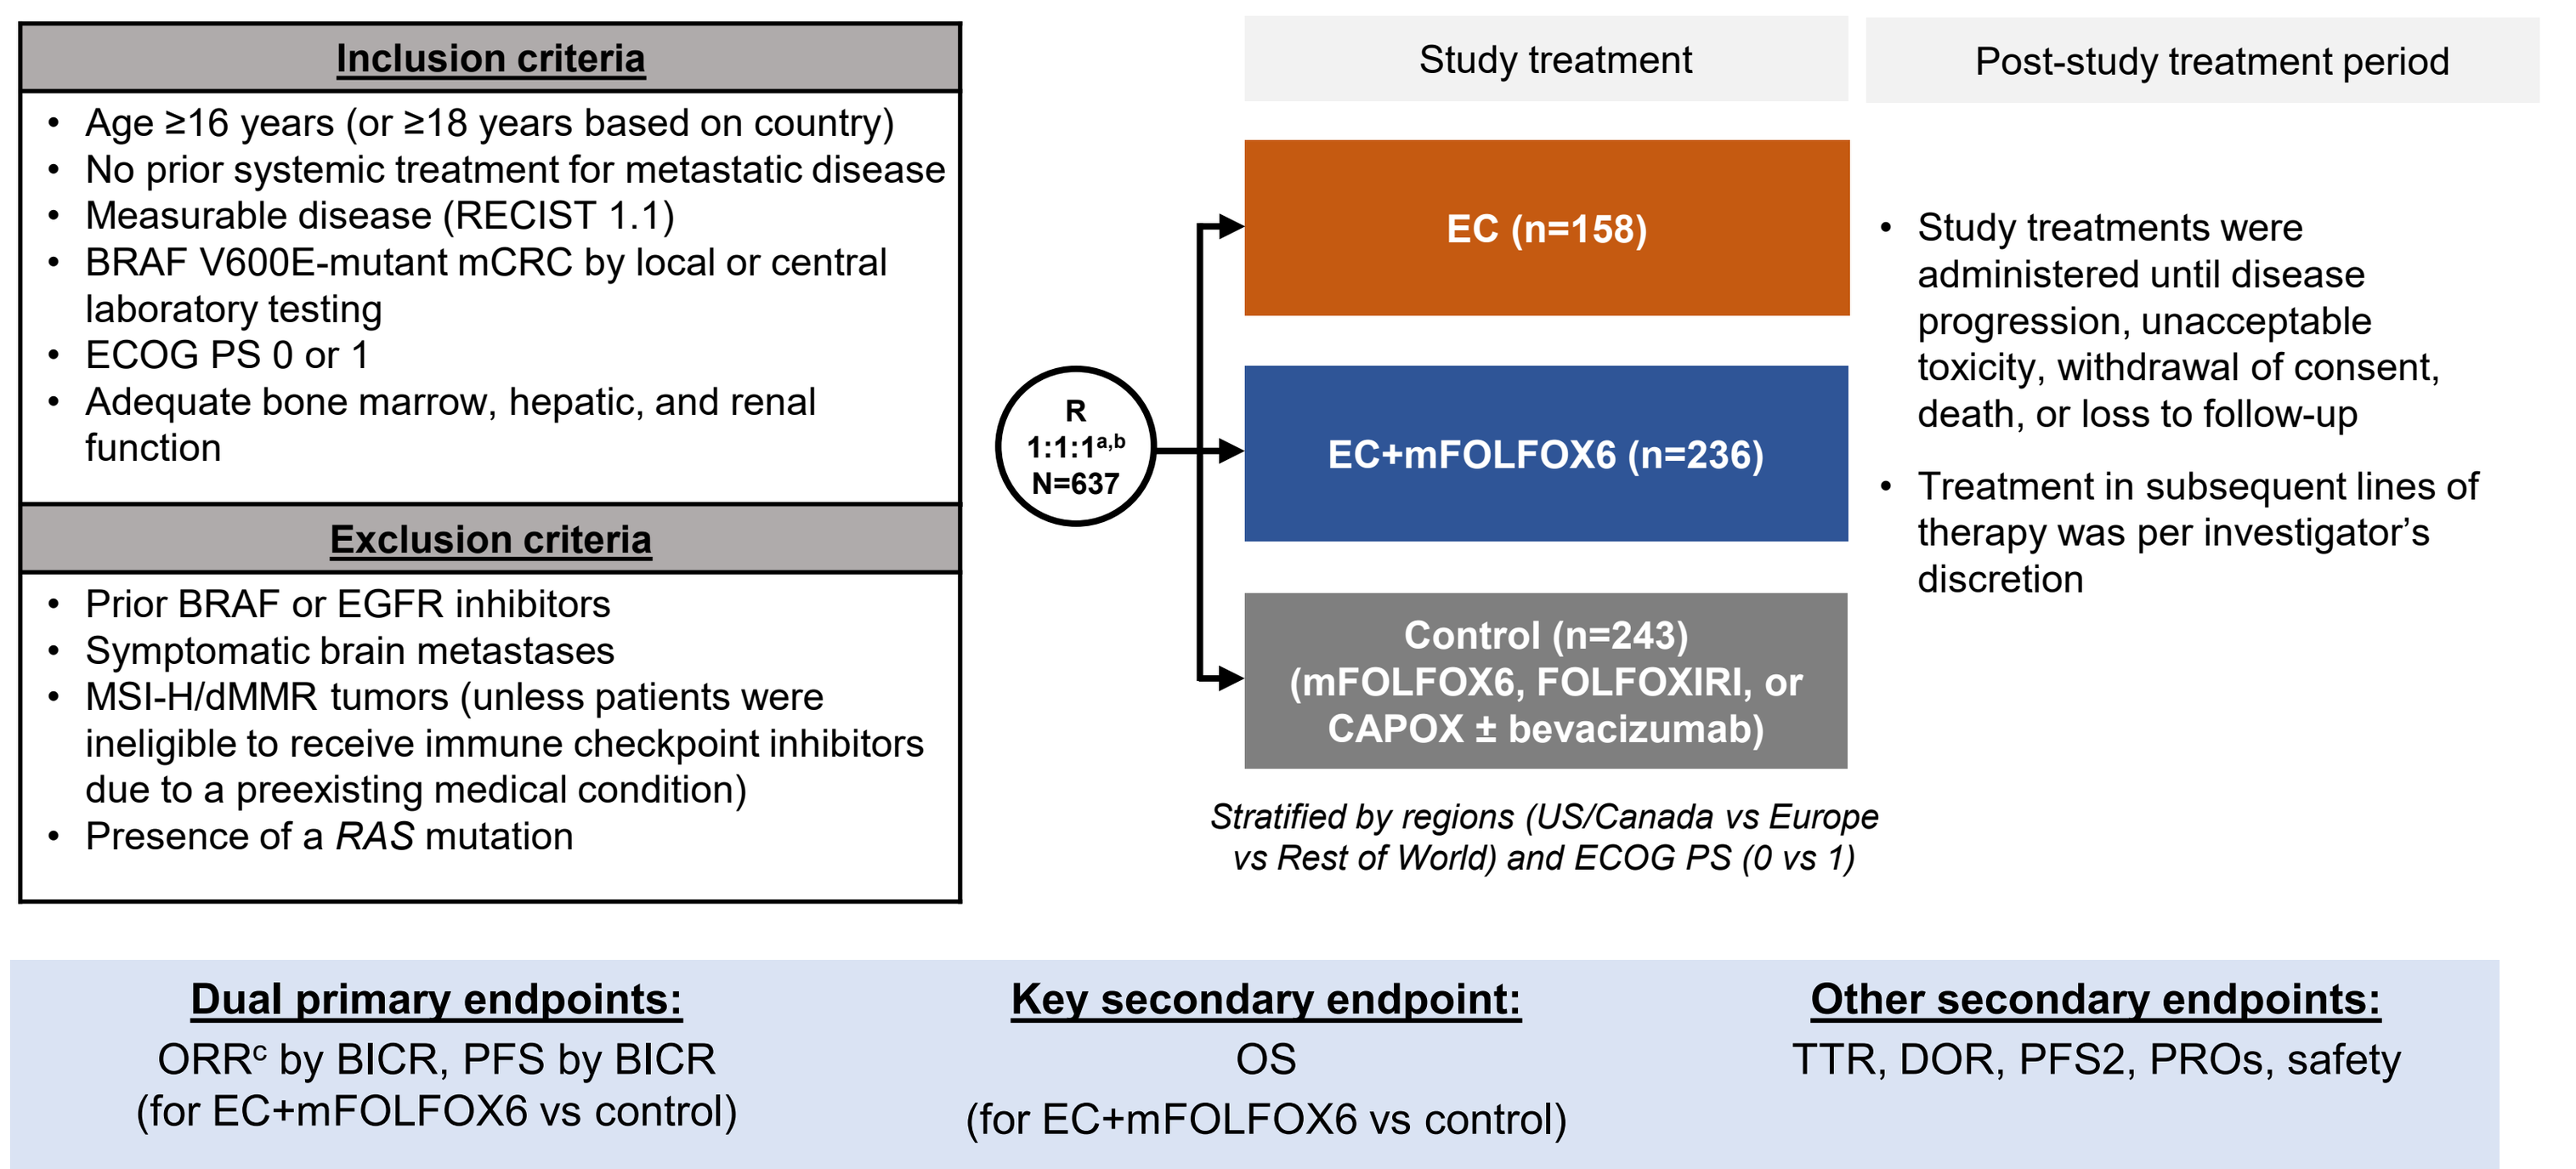

Inclusion criteria
Age ≥16 years (or ≥18 years based on country)
No prior systemic treatment for metastatic disease
Measurable disease (RECIST 1.1)
BRAF V600E-mutant mCRC by local or central laboratory testing
ECOG PS 0 or 1
Adequate bone marrow, hepatic, and renal function
Exclusion criteria
Prior BRAF or EGFR inhibitors
Symptomatic brain metastases
MSI-H/dMMR tumors (unless patients were ineligible to receive immune checkpoint inhibitors due to a preexisting medical condition)
Presence of a RAS mutation
R 1:1:1a,b N=637
Study treatment
Post-study treatment period
EC (n=158)
EC+mFOLFOX6 (n=236)
Control (n=243) (mFOLFOX6, FOLFOXIRI, or CAPOX ± bevacizumab)
Study treatments were administered until disease progression, unacceptable toxicity, withdrawal of consent, death, or loss to follow-up
Treatment in subsequent lines of therapy was per investigator's discretion
Stratified by regions (US/Canada vs Europe vs Rest of World) and ECOG PS (0 vs 1)
Dual primary endpoints:
ORRc by BICR, PFS by BICR (for EC+mFOLFOX6 vs control)
Key secondary endpoint:
OS (for EC+mFOLFOX6 vs control)
Other secondary endpoints:
TTR, DOR, PFS2, PROs, safety

[a]Following a protocol amendment, enrollment to the EC arm was stopped and patients were randomized 1:1 to the EC+mFOLFOX6 or control arms. [b]Patients were enrolled between November 16, 2021, and December 22, 2023. [c]In the first 110 patients in each of the EC+mFOLFOX6 and control arms.

Results

- The BREAKWATER EC, EC+mFOLFOX6, and control arms included 158, 236, and 243 patients, respectively, for the full analysis set and 153, 232, and 229 patients, respectively, for the safety analysis set, which consisted of all patients who received ≥1 dose of study treatment
- Baseline demographic and disease characteristics for the full analysis set were generally balanced between treatment groups[9]

Surgical conversion rate

- The number of patients who had cancer surgery with curative intent is shown in **Table 1:**

Table 1. Surgical Conversion Rate[a] (Safety Analysis Set)

n (%)	EC n=153	EC+mFOLFOX6 n=232	Control n=229
Patients with cancer surgery with curative intent	4 (2.6)	12 (5.2)	12 (5.2)
Patients with complete resection	3 (2.0)	7 (3.0)	8 (3.5)

[a]The percentage of patients who become eligible for surgery and undergo surgery with curative intent as a result of receiving study treatment.

Subsequent systemic anticancer therapies

- At the data cutoff of January 6, 2025, the majority of patients had discontinued study treatment. Of these patients, 73.3% in the EC arm, 63.9% in the EC+mFOLFOX6 arm, and 61.2% in the control arm received ≥1 subsequent systemic anticancer treatment (**Table 2**)
- Of the patients treated with subsequent systemic anticancer therapy, 19.6% (21/107), 17.6% (19/108), and 71.9% (100/139) in the EC, EC+mFOLFOX6, and control arms, respectively, received subsequent BRAF inhibitor-based therapies (**Table 2**)
 - The majority of patients in the EC and the EC+mFOLFOX6 arms received subsequent chemotherapies, with FOLFOX- and FOLFIRI-based regimens being the most commonly used in the EC arm and FOLFIRI-based regimens being the most commonly used in the EC+mFOLFOX6 arm
- Among patients who received a subsequent BRAF-inhibitor-based therapy in the control arm (n=100), most were treated with EC-based regimens (n=77) (**Table 2**)

Table 2. Subsequent Systemic Anticancer Therapies

Patients, n/N (%)	EC n=158	EC+mFOLFOX6 n=236	Control n=243
Discontinued study treatment	146/158 (92.4)	169/236 (71.6)	227/243 (93.4)
Any subsequent systemic anticancer treatment	107/146 (73.3)	108/169 (63.9)	139/227 (61.2)
Any BRAF inhibitor ± combination[a]	21/107 (19.6)	19/108 (17.6)	100/139 (71.9)
EC ± combination[a]	14/107 (13.1)	15/108 (13.9)	77/139 (55.4)
EC	4/107 (3.7)	9/108 (8.3)	62/139 (44.6)
Encorafenib + panitumumab ± combination[a]	0	1/108 (0.9)	4/139 (2.9)
Encorafenib + panitumumab	0	0	4/139 (2.9)

[a]Treatment ± combination denotes the indicated treatment given with or without any additional anticancer agents.

- In the control arm, subsequent BRAF inhibitor–based therapies predominated in the 2L, with 59.7% of patients (83/139) receiving BRAF inhibitor–based regimens vs 35.3% of patients (49/139) receiving chemotherapy–based regimens (**Table 3**)
 - Of the 83 patients who received a subsequent BRAF inhibitor–based therapy in the 2L, 53 patients received EC
- Most of the patients treated with BRAF inhibitor–based therapies in the control arm received these in the 2L (83%, 83/100) and/or 3L (18%, 18/100) of therapy (**Table 3**)

Table 3. Subsequent Systemic Anticancer Treatment by Line of Therapy

Patients receiving subsequent systemic anticancer treatment , n/N (%)	EC n=158	EC+mFOLFOX6 n=236	Control n=243
Patients who discontinued treatment	146/158 (92.4)	169/236 (71.6)	227/243 (93.4)
2L	107/146 (73.3)	108/169 (63.9)	139/227 (61.2)
BRAF inhibitor ± combination[a]	8/107 (7.5)	10/108 (9.3)	83/139 (59.7)
Chemotherapy-based[b]	99/107 (92.5)	91/108 (84.3)	49/139 (35.3)
Fruquintinib, regorafenib, or trifluridine/tipiracil ± VEGF inhibitor	0	2/108 (1.9)	2/139 (1.4)
Immunotherapy or other	0	5/108 (4.6)	5/139 (3.6)
3L	48/146 (32.9)	30/169 (17.8)	53/227 (23.3)
BRAF inhibitor ± combination[a]	8/48 (16.7)	10/30 (33.3)	18/53 (34.0)
Chemotherapy-based[b]	34/48 (70.8)	8/30 (26.7)	27/53 (50.9)
Fruquintinib, regorafenib, or trifluridine/tipiracil ± VEGF inhibitor	5/48 (10.4)	9/30 (30.0)	5/53 (9.4)
Immunotherapy or other	1/48 (2.1)	3/30 (10.0)	3/53 (5.7)
4L	21/146 (14.4)	7/169 (4.1)	18/227 (7.9)
BRAF inhibitor ± combination[a]	6/21 (28.6)	1/7 (14.3)	5/18 (27.8)
Chemotherapy-based[b]	7/21 (33.3)	3/7 (42.9)	7/18 (38.9)
Fruquintinib, regorafenib, or trifluridine/tipiracil ± VEGF inhibitor	7/21 (33.3)	2/7 (28.6)	6/18 (33.3)
Immunotherapy or other	1/21 (4.8)	1/7 (14.3)	0
5L	9/146 (6.2)	3/169 (1.8)	10/227 (4.4)
BRAF inhibitor ± combination[a]	1/9 (11.1)	2/3 (66.7)	1/10 (10.0)
Chemotherapy-based[b]	5/9 (55.6)	0	3/10 (30.0)
Fruquintinib, regorafenib, or trifluridine/tipiracil ± VEGF inhibitor	3/9 (33.3)	1/3 (33.3)	5/10 (50.0)
Immunotherapy or other	0	0	1/10 (10.0)
6L	6/146 (4.1)	0	4/227 (1.8)
BRAF inhibitor ± combination[a]	1/6 (16.7)	0	0
Chemotherapy-based[b]	2/6 (33.3)	0	1/4 (25.0)
Fruquintinib, regorafenib, or trifluridine/tipiracil ± VEGF inhibitor	3/6 (50.0)	0	3/4 (75.0)
7L	2/146 (1.4)	0	1/227 (0.4)
BRAF inhibitor ± combination[a]	0	0	0
Chemotherapy-based[b]	2/2 (100)	0	0
Immunotherapy or other	0	0	1/1 (100.0)
BRAF inhibitor ± combination[a] in any line	21/107 (19.6)	19/108 (17.6)	100/139 (71.9)
BRAF inhibitor ± combination[a] in >1 subsequent line	3/107 (2.8)	3/108 (2.8)	6/139 (4.3)

[a]Treatment ± combination denotes the indicated treatment given with or without any additional anticancer agents.
[b]CAPIRI ± combination, CAPOX ± combination, FOLFIRI ± combination, FOLFOX ± combination, FOLFOXIRI ± combination, or single-agent chemotherapy.

PFS2

- Median time from randomization to second progression, discontinuation of next-line treatment, or death was 20.7 months for patients in the EC+mFOLFOX6 arm compared with 12.9 months for patients in the control arm who received EC as their first subsequent treatment (**Figure 2**)

Figure 2. PFS2[a]

	EC+mFOLFOX6 n=236	Control with 2L EC (n=53)
Events	109 (46.2%)	43 (81.1%)
Median (95% CI)	20.7 months (19.0, 23.9)	12.9 months (11.2, 16.5)

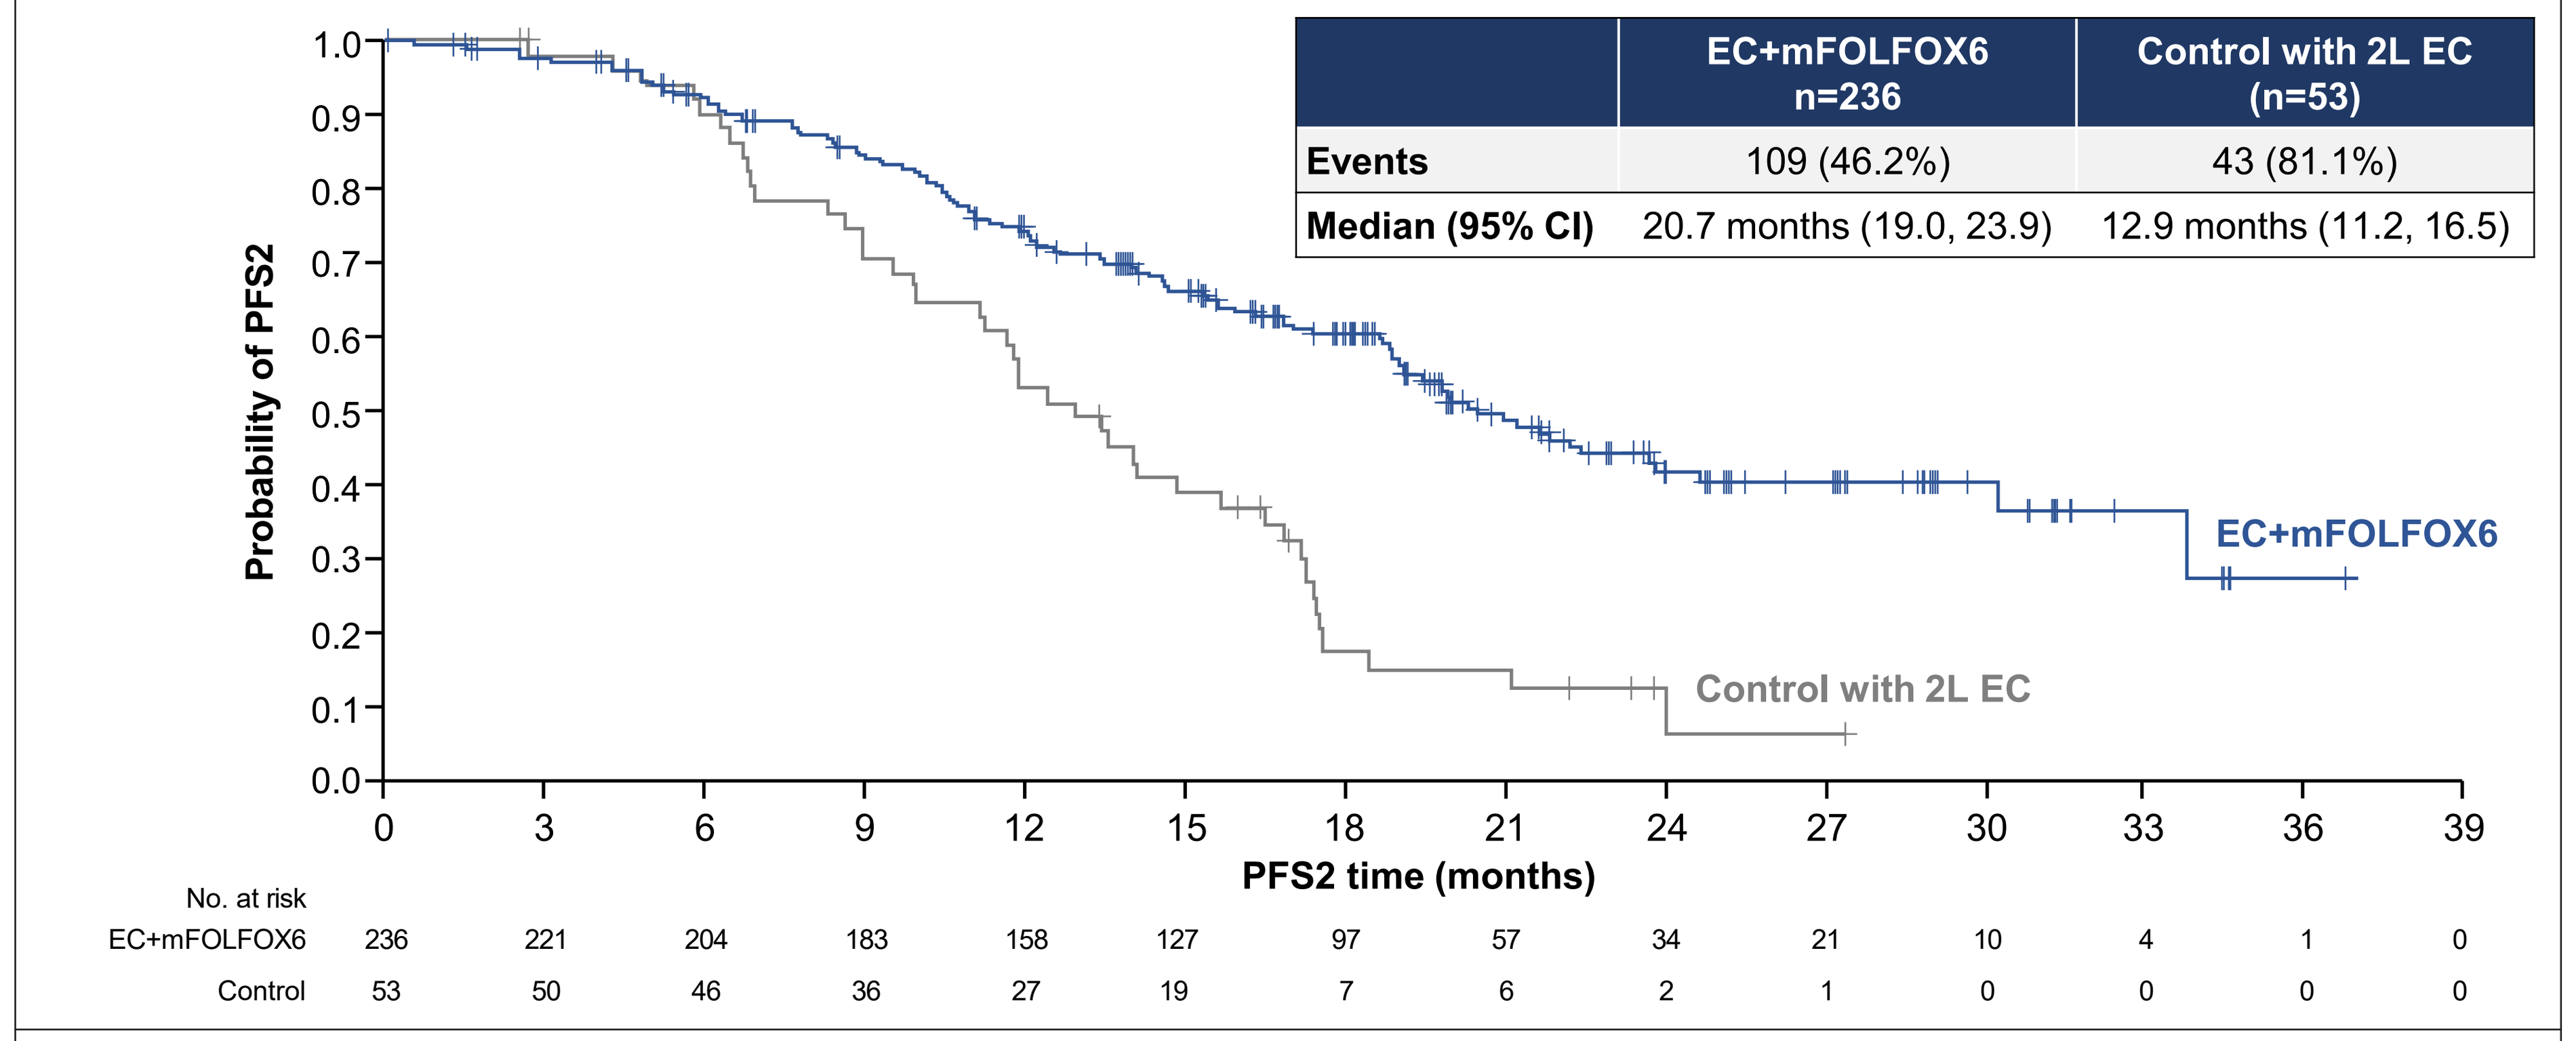

Probability of PFS2
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
EC+mFOLFOX6
Control with 2L EC
0 3 6 9 12 15 18 21 24 27 30 33 36 39
PFS2 time (months)
No. at risk
EC+mFOLFOX6 236 221 204 183 158 127 97 57 34 21 10 4 1 0
Control 53 50 46 36 27 19 7 6 2 1 0 0 0 0

[a]Time from randomization to second objective disease progression (progressive disease after the start of subsequent anticancer therapy), discontinuation of next-line treatment after first objective disease progression, or death from any cause, whichever occurs first.